Abstract No: 4611

Protection from Taxol- and Cisplatin-Induced Peripheral Neuropathy by AEG3482, a JNK Pathway Inhibitor

Peter Winocour, Kimberley Hewitt, Danielle Boulais, Panagiota Georgas, Catherine Cowan, Sandra Larouche, Jonathan Cole, Julie Johnstone, Nathalie Earl, Chris Simard, Christelle Pomies, Guillaume Levesque, Genevieve Doucet, Stephen Morris, James Jaquith, John Gillard, & Jon Durkin. Aegera Therapeutics, Inc., Montreal, Quebec, Canada

INTRODUCTION

Peripheral neuropathy is a dose-limiting side effect of both cisplatin and paclitaxel, which are used to treat a variety of cancers. Effective neuroprotective compounds have the potential to offer great clinical benefit, and increase the flexibility and intensity of dosing regimens for cancer patients. AEG3482 is a prototype of a family of compounds that prevents the activation of the JNK pathway, a crucial requirement for triggering neuronal apoptotic death from diverse insults, including chemotherapeutic drugs. AEG3482 was initially identified from an NGF withdrawal assay in SCG neurons, and subsequently screened in paclitaxel and cisplatin toxicity assays (Figure 1). Consistent with the role of the JNK pathway in apoptotic neuronal death, AEG3482 has proved to be a broad spectrum neuroprotective compound, and based on these characteristics, we predicted that AEG3482 could attenuate the damage to peripheral nerves resulting from chemotherapy-induced neuropathy. We, therefore, assessed the ability of AEG3482 to attenuate paclitaxel- and cisplatin-induced neuropathies in rat models that closely resemble the clinical situation.

METHODS

In vitro: Superior cervical ganglia (SCGs) were removed from post-natal day 1 rat pups. Ganglia were dissociated with trypsin and SCG neurons were plated in 96-well plates at a density of 8000 cells/well. Cells were cultured in Ultraculture (Biowitaker) supplemented with 3% rat serum, penicillin/streptomycin, L-glutamine, AraC, and 50 ng/ml NGF. After 5 days the media was replaced with fresh media (lacking rat serum and AraC) containing test compounds. Cells were exposed to chemotherapeutic agents and AEG3482 for 48 hrs. Cell viability was assessed by adding MTS or MTT to the media and incubating the cells for 4 hrs. MTT reduction was assessed by photomicrography. MTS reduction was assessed by measuring absorbance of the reduced dye at 570 nM.

In vivo paclitaxel: Rats were given paclitaxel twice a week (Monday & Thursday) (7 mg/kg, ip) for a total of five injections. AEG3482 (10 mg/kg, sc) was administered 1 hr prior to each paclitaxel injection. Using a quantitative method for analyzing functional gait disturbance with digital image-capture of paw prints, and H-reflex amplitude from direct sciatic nerve stimulation, we examined the neuroprotective effect of AEG3482 on paclitaxel-induced neuropathy.

In vivo cisplatin: Cisplatin was given to rats at 2.5 mg/kg, ip, daily for 5 consecutive days. AEG3482 was delivered (10 mg/kg, sc) 1 hr prior to, or 6 hr after each cisplatin injection. Sensory nerve conduction velocity (SNCV) was measured using the caudal nerve of the tail. SNCV was assessed prior to drug administration, and repeated 1, 2, and 3 weeks after the final cisplatin delivery. The effects of AEG3482 on cisplatin-induced expression of cyclin D1 in excised dorsal root ganglia (DRGs) was also examined. Tissues were dissected from animals receiving a cumulative dose of 15 mg/kg cisplatin over two delivery cycles, separated by one week, and changes in cyclin D1 protein levels were measured by Western blot.

COMPOUND SELECTION

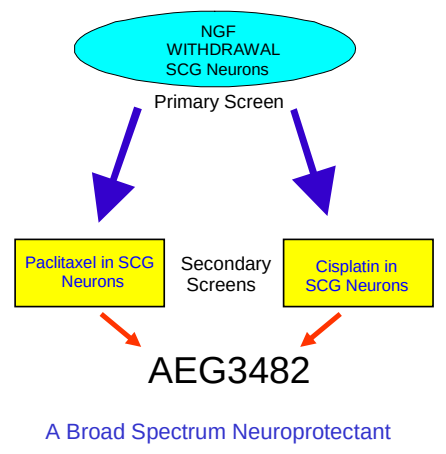
NGF WITHDRAWAL SCG Neurons
Primary Screen
Paclitaxel in SCG Neurons
Secondary Screens
Cisplatin in SCG Neurons
AEG3482
A Broad Spectrum Neuroprotectant

FIGURE 1: Compound selection criteria included an ability to rescue from NGF withdrawal, paclitaxel and cisplatin in vitro, and to have acceptable pharmacokinetic stability and bioavailability prior to testing in an *in vivo* neuropathy model.

RESULTS

AEG3482 Rescues Cultured SCG Neurons From NGF Withdrawal, Paclitaxel and Cisplatin (Figures 2-5)

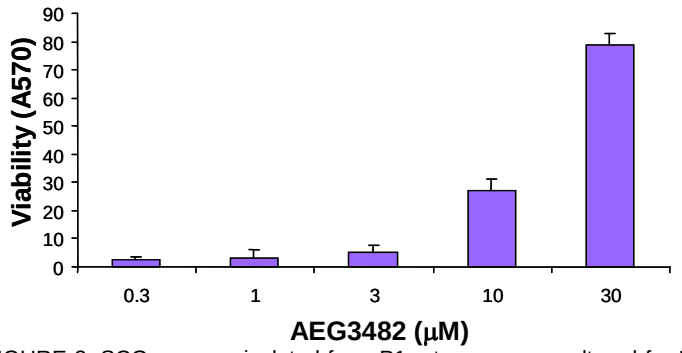
Viability (A570)
0.3
1
3
10
30
AEG3482 (μM)

FIGURE 2: SCG neurons isolated from P1 rat pups were cultured for 5 days in vitro as described in Methods.Trophic support was withdrawn by the addition of an anti-NGF polyclonal antibody to the culture media. Cell survival was assessed by MTT assay after 48 hrs of treatment.

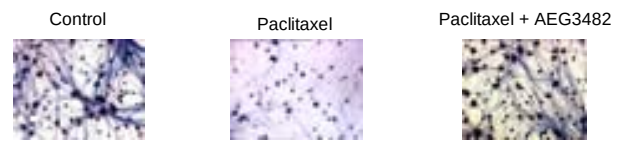
Control
Paclitaxel
Paclitaxel + AEG3482

FIGURE 3: AEG3482 maintains neuritic metabolism in the presence of paclitaxel. SCG neurons were cultured from P1 rat pups as described in Methods. Cells were either untreated (Control), treated with 300 ng/ml paclitaxel or 300 ng/ml paclitaxel and 30 μM 3482. After 48 hrs MTT was added to the culture media and cellular accumulation of biotransformed MTT was assessed. Paclitaxel specifically prevented MTT reduction in neurites. AEG3482 treatment completely protects neurites from paclitaxel.

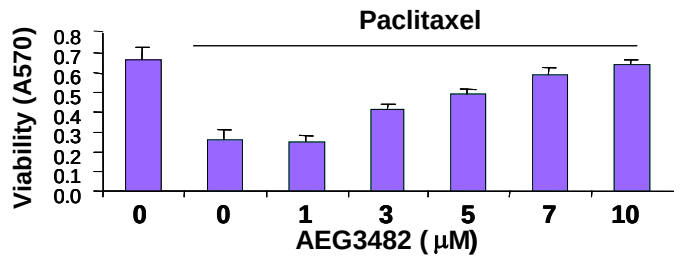
Paclitaxel
Viability (A570)
0 0 1 3 5 7 10
AEG3482 (μM)

FIGURE 4: AEG3482 maintains SCG neuron viability in the presence of paclitaxel. SCG neurons cultured from P1 rat pups were exposed to 300 ng/ml paclitaxel in the presence of the indicated concentrations of AEG3482. After 48 hrs MTS dye was added to the media and reduction of MTS, which correlated with viability, was determined. AEG3482 protected SCG neurons from paclitaxel in a dose responsive manner.

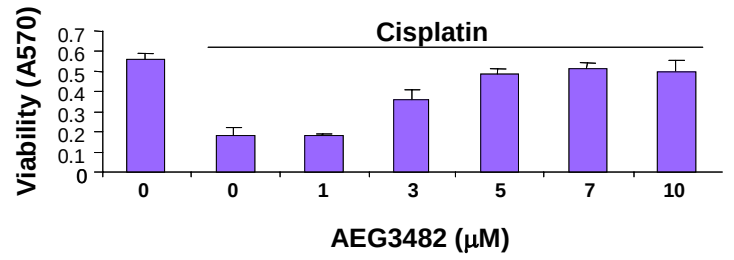
Cisplatin
Viability (A570)
0 0 1 3 5 7 10
AEG3482 (μM)

FIGURE 5: SCG neurons from P1 rat pups were exposed to 2.5 μg/ml cisplatin in the presence of the indicated concentrations of AEG3482. After 48 hrs MTS dye was added to the media and the reduction of MTS was determined. AEG3482 protected SCG neurons from cisplatin in a dose responsive manner.

AEG3482 Inhibits the JNK pathway

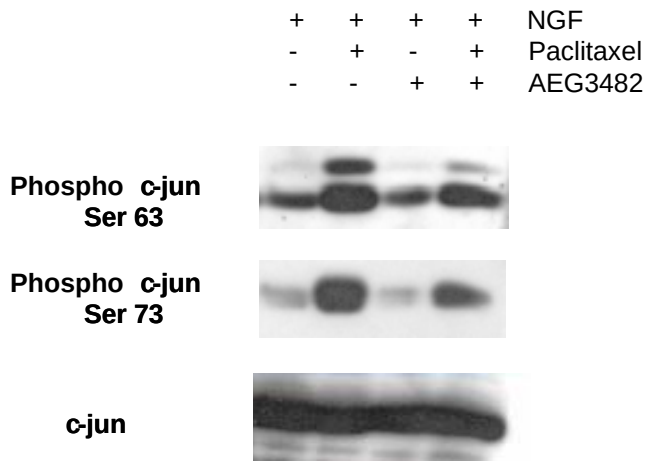
+ + + + NGF
- + - + Paclitaxel
- - + + AEG3482
Phospho c-jun Ser 63
Phospho c-jun Ser 73
c-jun

FIGURE 6: SCG neurons from P1 rat pups were exposed to paclitaxel (300 ng/ml) and/or AEG3482 (30 μM) for 24 hrs. Cell lysates were ran on SDS-polyacrylamide gels, transferred to nitrocellulose and antibodies specific for c-jun and phospho-c-jun (ser 63 & 73) used to detect the total and JNK-activated forms.

AEG3482 Does Not Rescue H460 Lung Cancer Cells from Chemotherapy

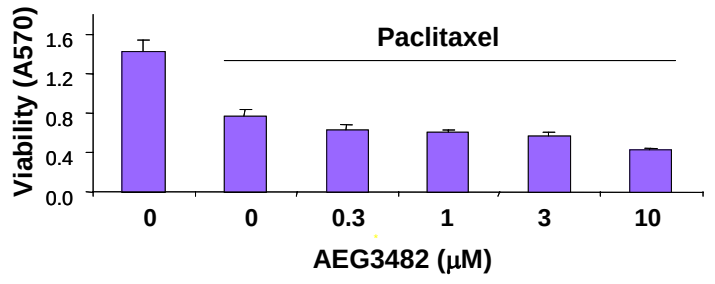
Paclitaxel
Viability (A570)
0 0 0.3 1 3 10
AEG3482 (μM)

FIGURE 7: While rescuing neurons, AEG3482 does not protect H460 tumor cells from paclitaxel. Human non-small cell lung carcinoma cells (H460) were treated with 300 ng/ml paclitaxel. Cells were co-treated with AEG3482 at the indicated concentrations for 48 hrs. Cellular viability was assessed by MTS. This result is consistent with the differential role the JNK pathway plays in neuronal and cancer cell survival. These results are representative of the effect of AEG3482 on multiple cancer cell lines, supporting the use for AEG3482 as a protective agent against chemotherapeutic-induced neuropathy, without interfering with cancer treatment.

AEG3482 Prevents Paclitaxel-Induced Disruption of Normal Walking Ability

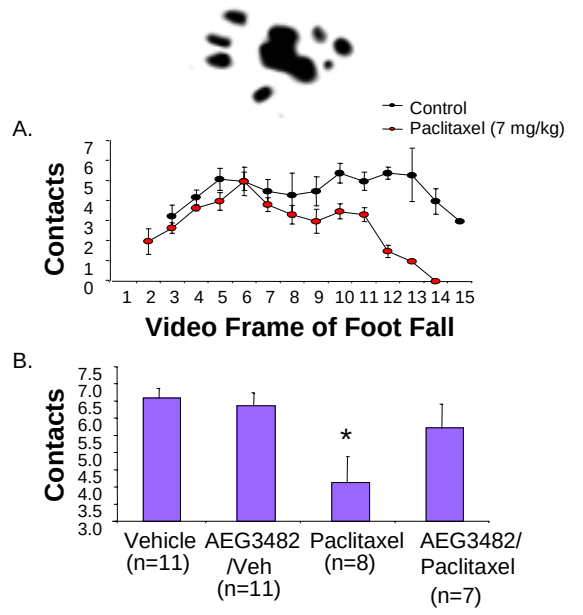
A.
Control
Paclitaxel (7 mg/kg)
Contacts
Video Frame of Foot Fall
B.
Contacts
Vehicle (n=11)
AEG3482 /Veh (n=11)
Paclitaxel (n=8)
AEG3482/ Paclitaxel (n=7)

FIGURE 8: (A) Paclitaxel administration to rats (as described in Methods) induced a "tip-toe" walking stance resulting in decreased contact in the most ventral portion of the back hindlimb during voluntary ambulation. (B) Administration of AEG3482 (10 mg/kg) 1 hr prior to each paclitaxel injection attenuated the effects of chemotherapy on functional walking ability. (* $p < 0.05$, significantly different from Vehicle alone).

AEG3482 Prevents Paclitaxel-Induced Attenuation of H-reflex Amplitude

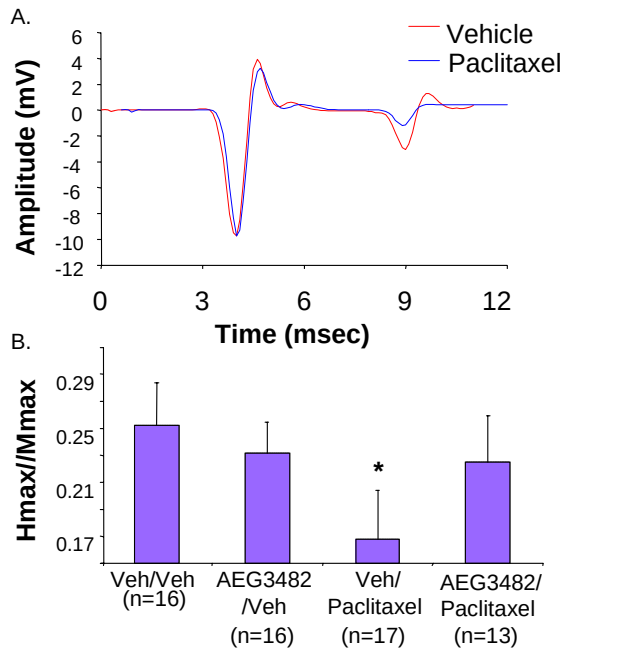
A.
Vehicle
Paclitaxel
Amplitude (mV)
Time (msec)
B.
Hmax//Mmax
Veh/Veh (n=16)
AEG3482 /Veh (n=16)
Veh/ Paclitaxel (n=17)
AEG3482/ Paclitaxel (n=13)

FIGURE 9: AEG3482 prevents the reduction in H-reflex amplitude resulting from paclitaxel-induced neuropathy. (A) Paclitaxel causes a decrease in H-reflex amplitude. (B) Using H-reflex as an indication of sensory nerve function, AEG3482 prevented the effects of paclitaxel. (* $p < 0.05$, significantly different from Veh/Veh).

AEG3482 Prevents Cisplatin-Induced Attenuation of Sensory Nerve Conduction Velocity (SNCV)

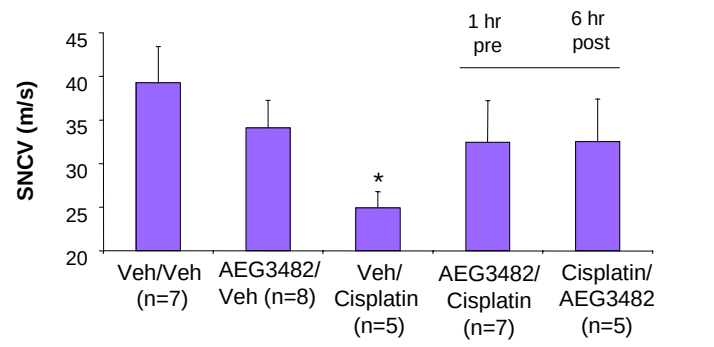
SNCV (m/s)
1 hr pre
6 hr post
Veh/Veh (n=7)
AEG3482/ Veh (n=8)
Veh/ Cisplatin (n=5)
AEG3482/ Cisplatin (n=7)
Cisplatin/ AEG3482 (n=5)

FIGURE 10: Cisplatin-induced neuropathy is attenuated by AEG3482 therapy either when administered 1 hr prior to or 6 hrs after each cisplatin injection. Cisplatin was delivered at a cumulative dose of 12.5 mg/kg, ip over 5 days. AEG3482 was given sc as described in Methods. SNCV was assessed two weeks after the final drug administration. (* $p < 0.05$, significantly different from Veh/Veh).

AEG3482 Reduces Cyclin D1 Expression in DRGs from Rats Treated with Cisplatin

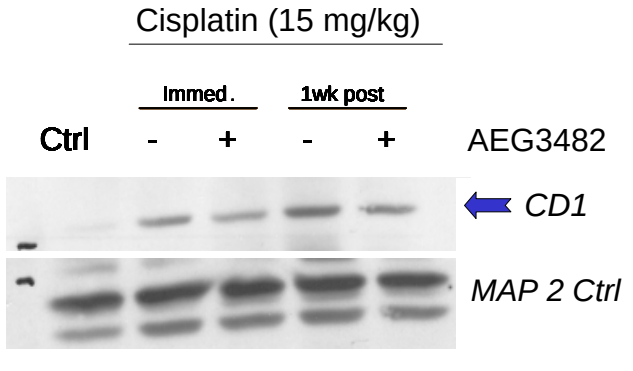
Cisplatin (15 mg/kg)
Immed.
1wk post
Ctrl - + - + AEG3482
CD1
MAP 2 Ctrl

FIGURE 11: Cyclin D1 expression was induced by cisplatin treatment when assessed on the final day of administration. This expression continued to increase over a 1 week period, despite cessation of chemotherapy. AEG3482 co-treatment partially attenuated the effect of cisplatin on cyclin D1 expression. MAP2 was used as a protein loading control.

CONCLUSIONS:

•AEG3482 was selected as a broad spectrum neuroprotectant with JNK pathway inhibitory properties.

•AEG3482 protects SCG neurons in vitro from NGF withdrawal, cisplatin and paclitaxel.

•AEG3482 effectively reduces chemotherapy-induced neuropathy in rats treated with paclitaxel, preventing the decrement in sensory nerve function (H-reflex) and functional walking ability.

•AEG3482 effectively prevents cisplatin-induced neuropathy, maintaining SNCV and reducing abnormal cyclin D1 expression.

•AEG3482 is a potent neuroprotective agent against chemotherapeutic-induced neuropathy and is currently under development for clinical use. As a broad-spectrum neuroprotectant that inhibits the JNK pathway, AEG3482 may have the ability to save neurons from a number of neurotoxic insults, such as those arising from diabetes, traumatic injury, or motor neurons disease (e.g., ALS, SMA). Compound testing in these models is currently underway.

1